Abstract Number: **3774** | Poster Board Number: **P466** | www.jrfglobal.com

DETERMINATION OF IMMUNOTOXIC POTENTIAL OF FENBUTATIN OXIDE IN HSD:ICR (CD-1®) MICE

Darpesh Gohel[1], Poonam P. Mehta[1], Sudhakar Jadhav[1], Kunjan Shah[1], Mehul Pandya[1]; Manish Patel[1] and Vincent J. Piccirillo[2]
[1]Department of Toxicology, Jai Research Foundation, Gujarat, India & [2] VJP Consulting, Inc., Ashburn, VA USA

JAI RESEARCH FOUNDATION

ABSTRACT

This study was conducted to determine the immunosuppressive effect of Fenbutatin Oxide (FBTO) by repeated daily oral administration via diet for 28 days in mice following guidelines of EPA OPPTS 870.7800.

Fifty mice of each sex were randomly divided into five groups (G1 to G5), 10 mice/sex/group. FBTO was given at 0 (G1, control; basal diet), 50 (G2), 150 (G3) and 450 (G4) ppm in diet. Mice from positive control group (G5) were treated with cyclophosphamide (CYM) at 50 mg/kg b. wt./day for a period of 4 consecutive days (Day 25-28) via intraperitoneal injection. All mice (G1-G5) were treated with sheep red blood cells (SRBC) in normal saline at fixed dose volume of 0.2 mL/mouse (7.8 x 10^7 RBCs) by intravenous injection on day 25.

Mice were observed twice for clinical signs, morbidity and mortality. Body weight and food consumption of individual mouse were determined twice weekly throughout the study. An ELISA was performed using Mouse Anti-SRBC IgM ELISA kit to determine the effects on serum anti-SRBC IgM levels. Animals were subjected to gross pathology and organ weight was determined for spleen and thymus. All mice were alive without showing any clinical sign during experiment. Marked reductions in body weight, food consumption and anti-SRBCs IgM level was observed for males and females at 450 ppm of FBOT. Statistically, significant reduction in absolute weights of spleen and thymus were observed in the group treated with 450 ppm FBOT which was similar to that of CYM treated group. External and internal examination of terminally sacrificed animals did not reveal any pathological lesions.

Based on the results, it was concluded that, FBOT produced immunosuppressive effects at 450 ppm dose level when administered orally through diet for 28 consecutive days in male and female mice of Hsd:ICR (CD-1®) strain. FBOT induced immunosuppression was comparable in both sexes.

INTRODUCTION TO STUDY PROCEDURE

Test Item	Fenbutatin Oxide Technical (FBTO)
Test System & Groups	Female Hsd:ICR (CD-1®) Mice Five groups (G1 - Vehicle Control, G2 - Low Dose, G3 - Mid Dose, G4 - High Dose and G5 - Positive control) 10 male and 10 female mice per Group
Guideline	EPA OPPTS 870.7800
Dose Levels	G1 - 0 ppm, G2 - 50 ppm, G3 - 150 ppm, and G4 - 450 ppm FBTO in diet; G5 - 50 mg/kg b. wt./day cyclophosphamide via intraperitoneal injection
Sheep red blood cells (SRBC)	All mice (G1-G5) were treated with SRBC at 0.2 mL/mouse (atleast 7.8 x 107 RBCs) by IV injection on day 25
Diet Analysis	Formulated diet was analysed for Stability, homogeneity and A.I. concentration analysis
Observations	Clinical signs, mortality and morbidity : Twice a day Detailed Physical Observation : Once a Week Body weight and food consumption : Twice a week serum anti-SRBC IgM estimation by ELISA Gross Pathology Absolute and relative organ weights for the spleen and thymus

Table 1 showing significant reduction in mean body weight of high dose treated male mice

Day	Vehicle Control		Low Dose		Mid Dose		High Dose		Positive Control	
	Mean	SD	Mean	SD	Mean	SD	Mean	SD	Mean	SD
15	38.98	3.58	39.42	1.95	38.29	2.81	35.49↓	3.68	38.35	1.58
18	39.57	3.79	39.88	2.28	38.89	2.85	35.73↓	3.26	38.68	1.83
22	40.26	4.14	40.49	2.32	38.47	2.90	35.11↓↓	3.29	39.06	2.36
25	40.50	4.46	40.67	2.42	39.11	2.73	36.12↓	3.41	39.07	2.32
28	41.05	4.46	41.18	2.40	38.60	2.98	36.17↓↓	3.55	39.07	2.40

Table 2 showing significant reduction in mean body weight of high dose treated female mice

Day	Vehicle Control		Low Dose		Mid Dose		High Dose		Positive Control	
	Mean	SD	Mean	SD	Mean	SD	Mean	SD	Mean	SD
18	27.30	2.65	25.87	1.44	26.03	1.65	24.51↓	1.57	26.93	1.69
22	27.88	1.63	26.29	1.44	26.43	2.13	24.70↓↓	1.75	27.74	1.94
25	28.23	1.91	26.53	1.35	26.60	1.91	25.10↓↓	1.61	28.33	2.00
28	28.13	1.62	26.36	1.22	26.74	2.00	24.54↓↓	1.39	28.09	2.15

Keys: ↓↓ - Significantly lower than control ($p \leq 0.01$); ↓ - Significantly lower than control ($p \leq 0.05$)

RESULTS

- ❖ No mortality was observed during the study period.
- ❖ All the animals were normal throughout the study period.
- ❖ No abnormalities were observed in animals from all the groups during detailed physical/clinical examinations.
- ❖ Treatment related marked reductions in mean body weight and feed consumption was observed in high dose (450 ppm) treated group.
- ❖ Significant reduction in Anti-SRBCs IgM antibodies production at high dose (450 ppm in diet) level
- ❖ Significant reduction in absolute weights of spleen and thymus at 450 ppm. The effect observed in absolute weight of spleen and thymus was similar to the effect observed in the cyclophosphamide treated positive control group.
- ❖ External and internal examination of terminally sacrificed animals did not reveal any lesions of pathological significance.

Table 3 showing significant reduction of Anti-SRBC IgM in high dose and positive control group

Vehicle Control		Low Dose		Mid Dose		High Dose		Positive Control	
Mean	SD	Mean	SD	Mean	SD	Mean	SD	Mean	SD
3675.98	34.03	4854.42	1157.17	4255.34	680.67	1034.92↓↓	125.77	20.86↓↓	48.14

Keys: ↓↓ - Significantly lower than control ($p \leq 0.01$); ↓ - Significantly lower than control ($p \leq 0.05$)

Figure 1 showing reduction in Anti-SRBC IgM concentration due to FBTO & CYP treatment

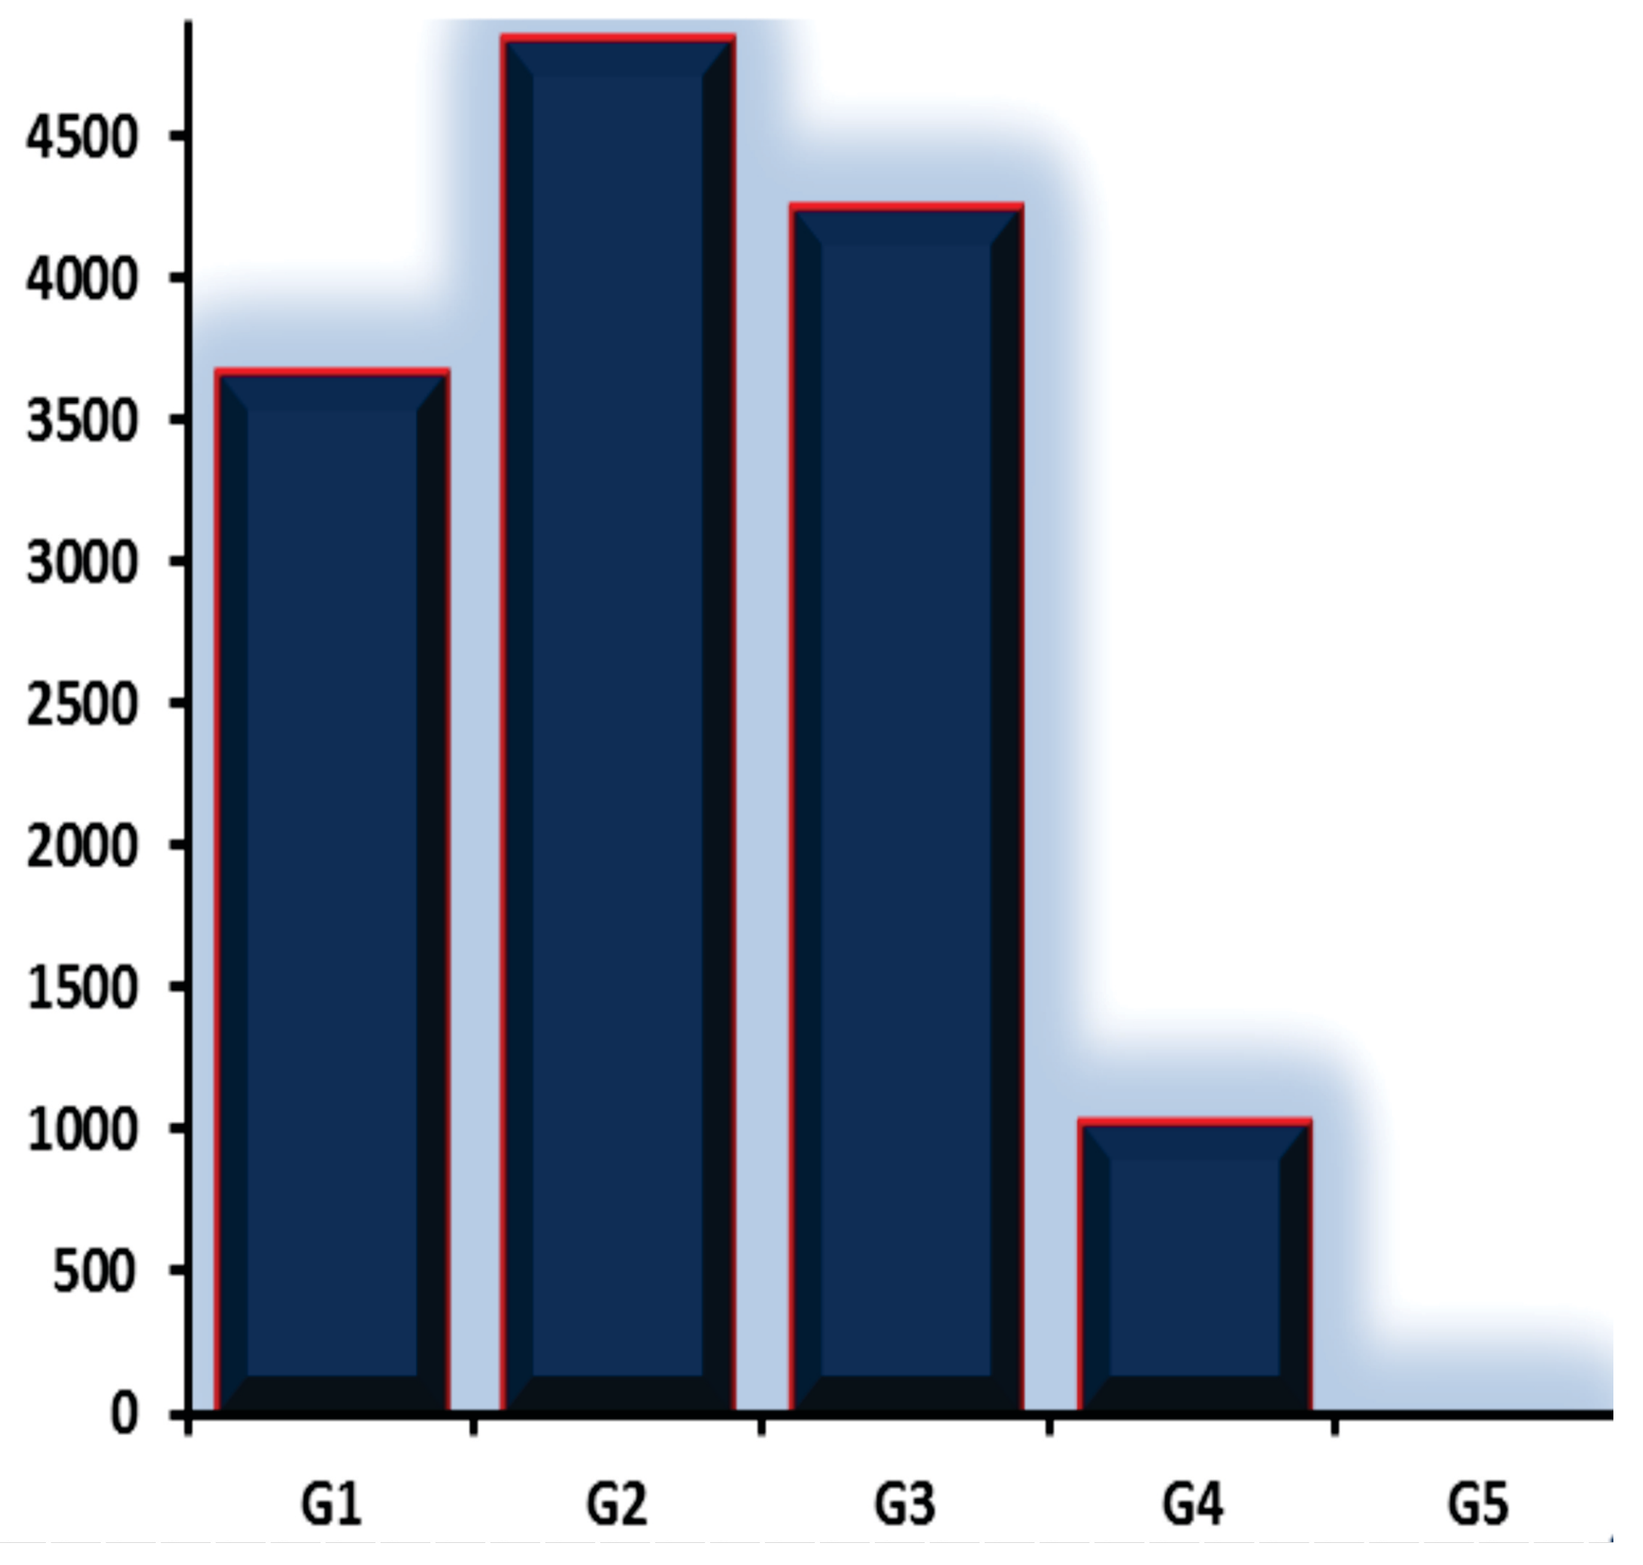
4500
4000
3500
3000
2500
2000
1500
1000
500
0
G1
G2
G3
G4
G5
Group No.

Figure 2: Showing significant reduction in mean feed consumption of high dose treated male mice

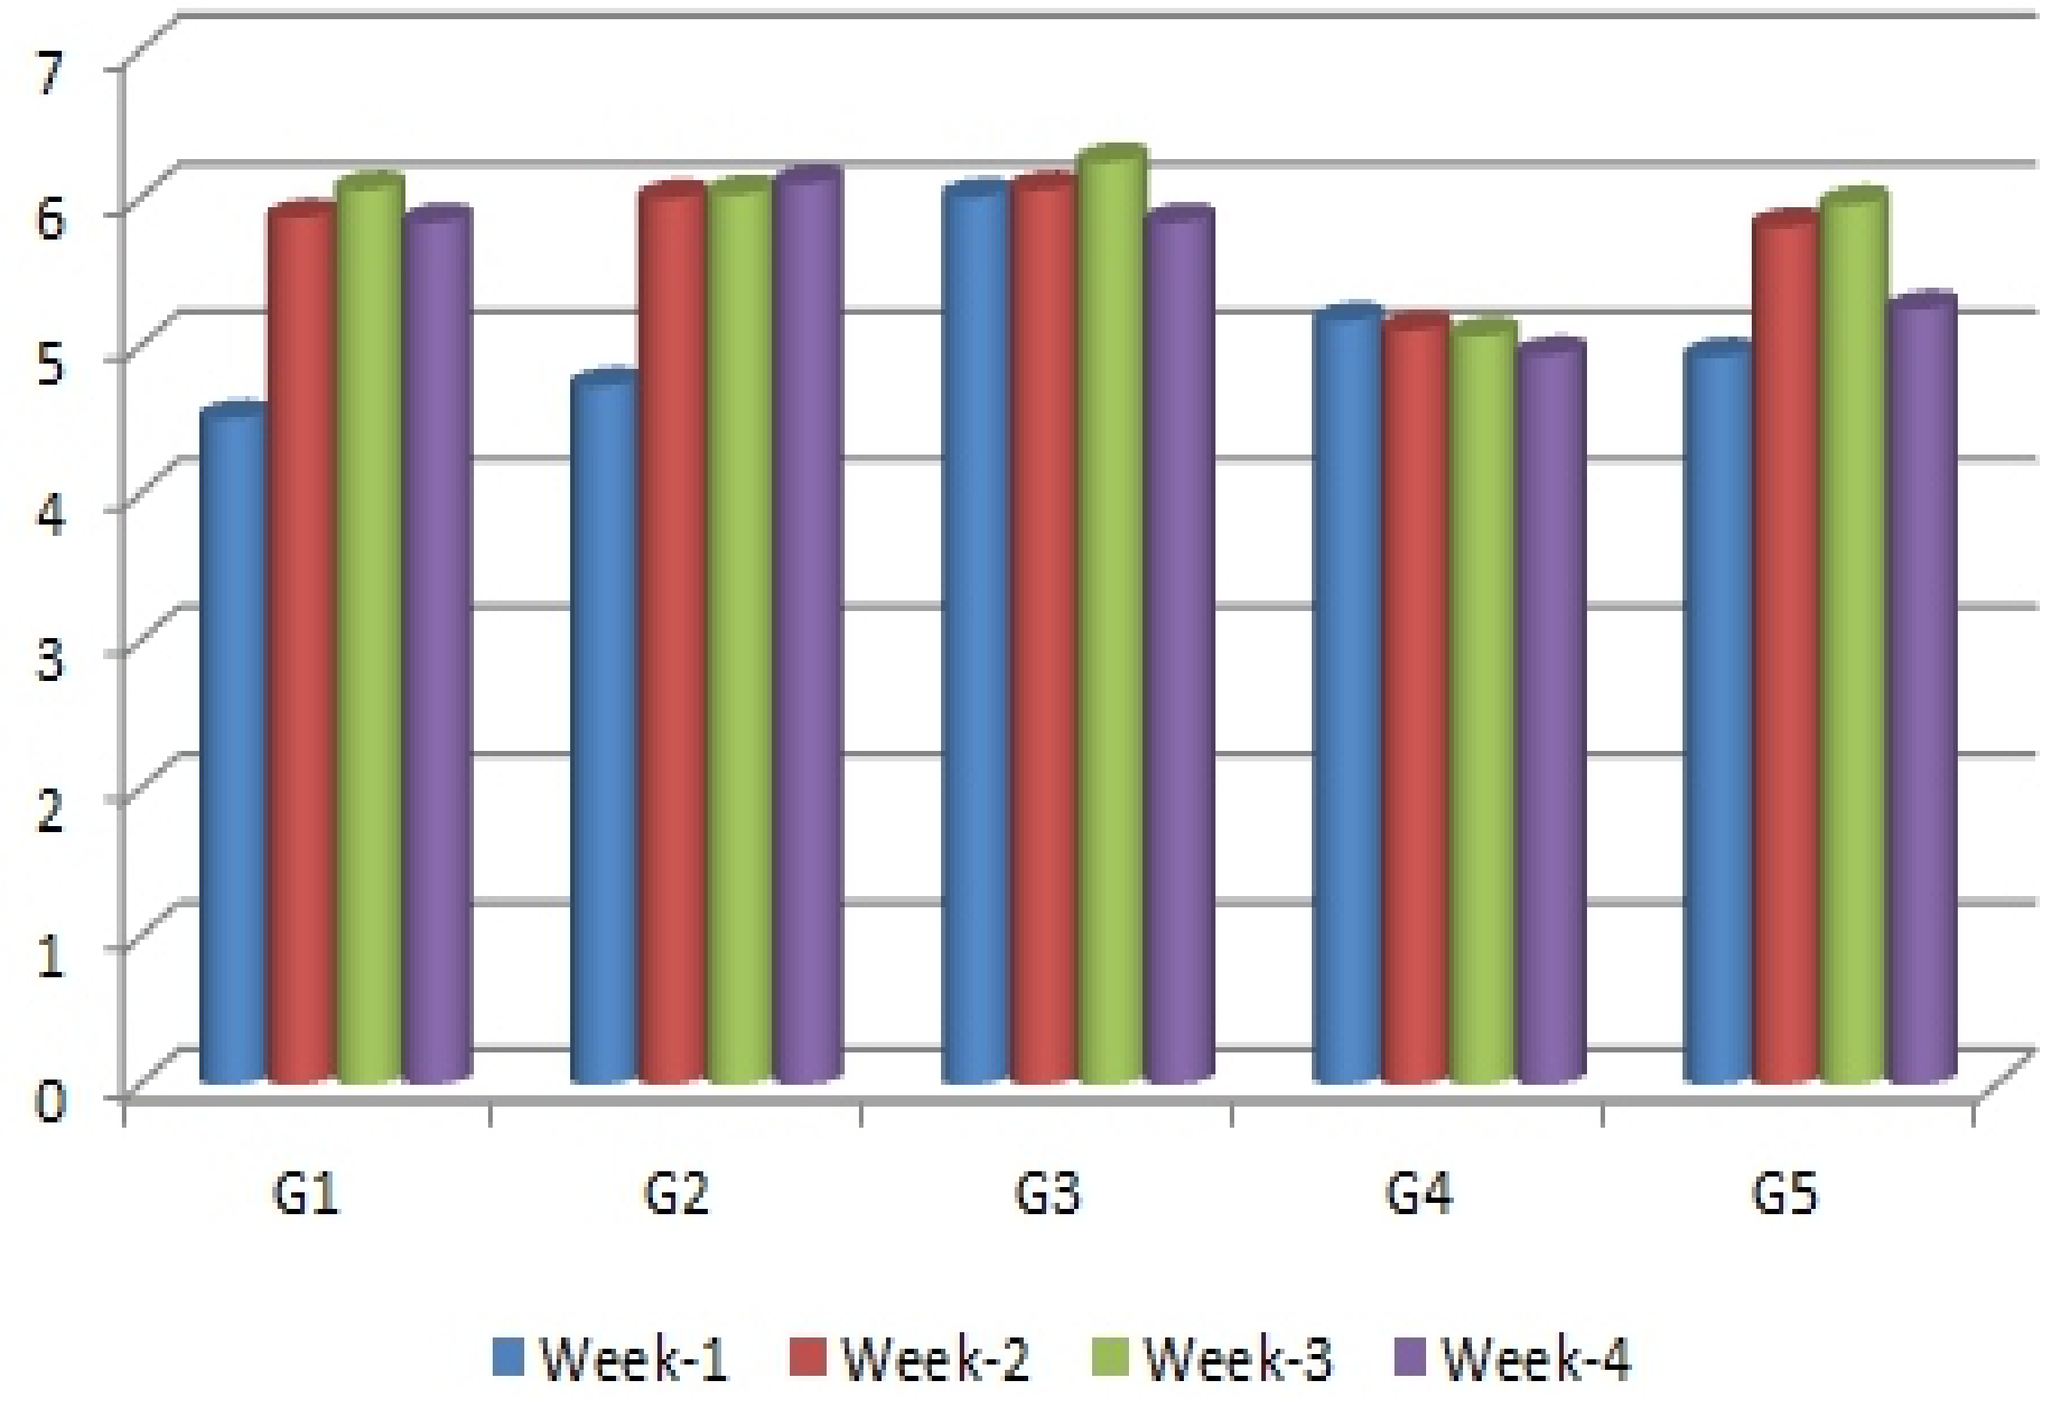
7
6
5
4
3
2
1
0
G1
G2
G3
G4
G5
Week-1
Week-2
Week-3
Week-4

Figure 3: Showing significant reduction in mean feed consumption of high dose treated female mice

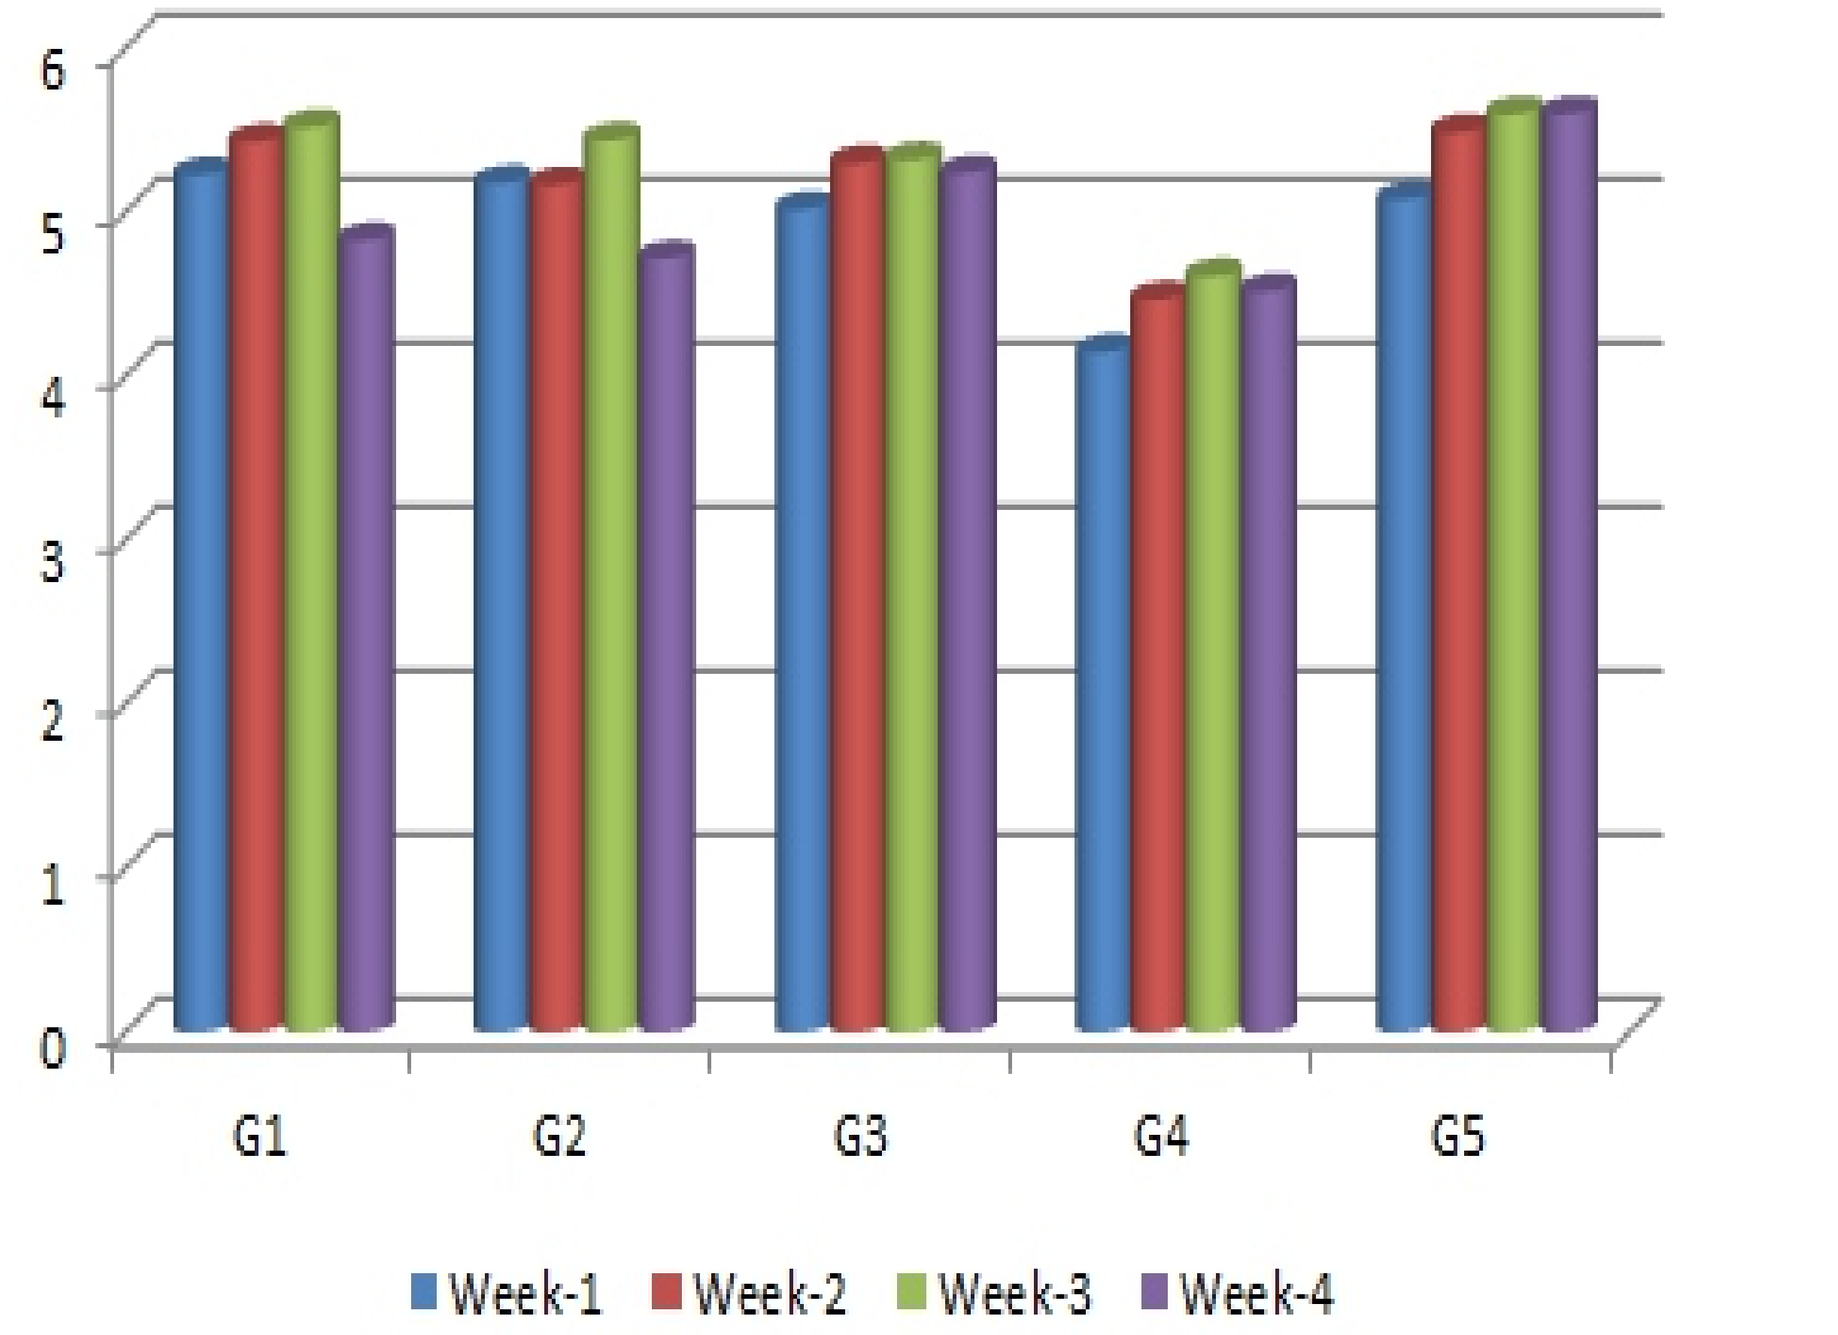
6
5
4
3
2
1
0
G1
G2
G3
G4
G5
Week-1
Week-2
Week-3
Week-4

CONCLUSION

Under the conditions of this study, it is concluded that Fenbutatin Oxide Technical produced immunosuppressive effects at the dose levels of 450 ppm in diet (equivalent to 64.43 ± 6.94 and 81.27 ± 5.40 mg/kg b.wt. for males and females, respectively), when administered orally through diet for 28 consecutive days in mice of Hsd:ICR (CD-1®) strain.